A novel integrated NMR-based approach for studying receptor-ligand interactions on living cells surface

C. Acconcia[‡], B. Farina[⊥], M. Gentile[‡], S. Di Gaetano[‖], D. Capasso[§], A. Del Gatto[‖], A. Paladino[ǀ], M. Saviano[ǂ], R. Fattorusso[‡], L. Zaccaro[‖] and L. Russo[‡].

[‡]Department of Environmental, Biological and Pharmaceutical Science and Technology, University of Campania - Luigi Vanvitelli, via Vivaldi 43, 81100 Caserta, Italy;
[⊥]Advanced Accelerator Applications, a Novartis Company, via Vivaldi 43, 81100 Caserta, Italy;
[‖]Institute of Biostructures and Bioimaging-CNR, via Mezzocannone 16, 80134, Naples, Italy;
[§]Interdepartmental Center of Bioactive Peptide, University of Naples Federico II, Via Mezzocannone 16, 80134 Naples, Italy;
[ǀ]Department of Science and Technology, University of Sannio, via Francesco de Sanctis, Benevento 82100, Italy;
[ǂ]Institute of Crystallography-CNR, via Amendola 122/O, 70126 Bari, Italy.

Introduction

Nuclear Magnetic Resonance Spectroscopy represents a powerful technique for studying, at atomic resolution, protein-protein and protein-ligand interactions, directly in the intracellular environment or on the membrane surface of living cells. In- and on-cell NMR methods as transfer NOESY (trNOESY) and Saturation Transfer Difference (STD), are widely used to characterize binding of ligands to membrane receptors on the cell surface. However, the application of both NMR methodologies is limited by the short lifetime of cells in an NMR sample tube, which often prevents the acquisition of experiments longer than few hours [1,2]. Therefore, to overcome these drawbacks, we developed an alternative approach based on the application of Non-Uniform Sampling (NUS) and 1D T1ρ NMR techniques to collect structural and dynamics information on the receptor-ligand interactions with living cells, that in turn can be used as conformational constraints in computational studies. We tested our approach to explore the recognition mechanism of αvβ5-integrin by RGDechi15D [3,4]. This peptide is a selective cyclic molecule able to interfere with tumor proliferation and progression and to regulate angiogenesis in endothelial cells.

Results

ps-ns dynamics of RGDechi15D

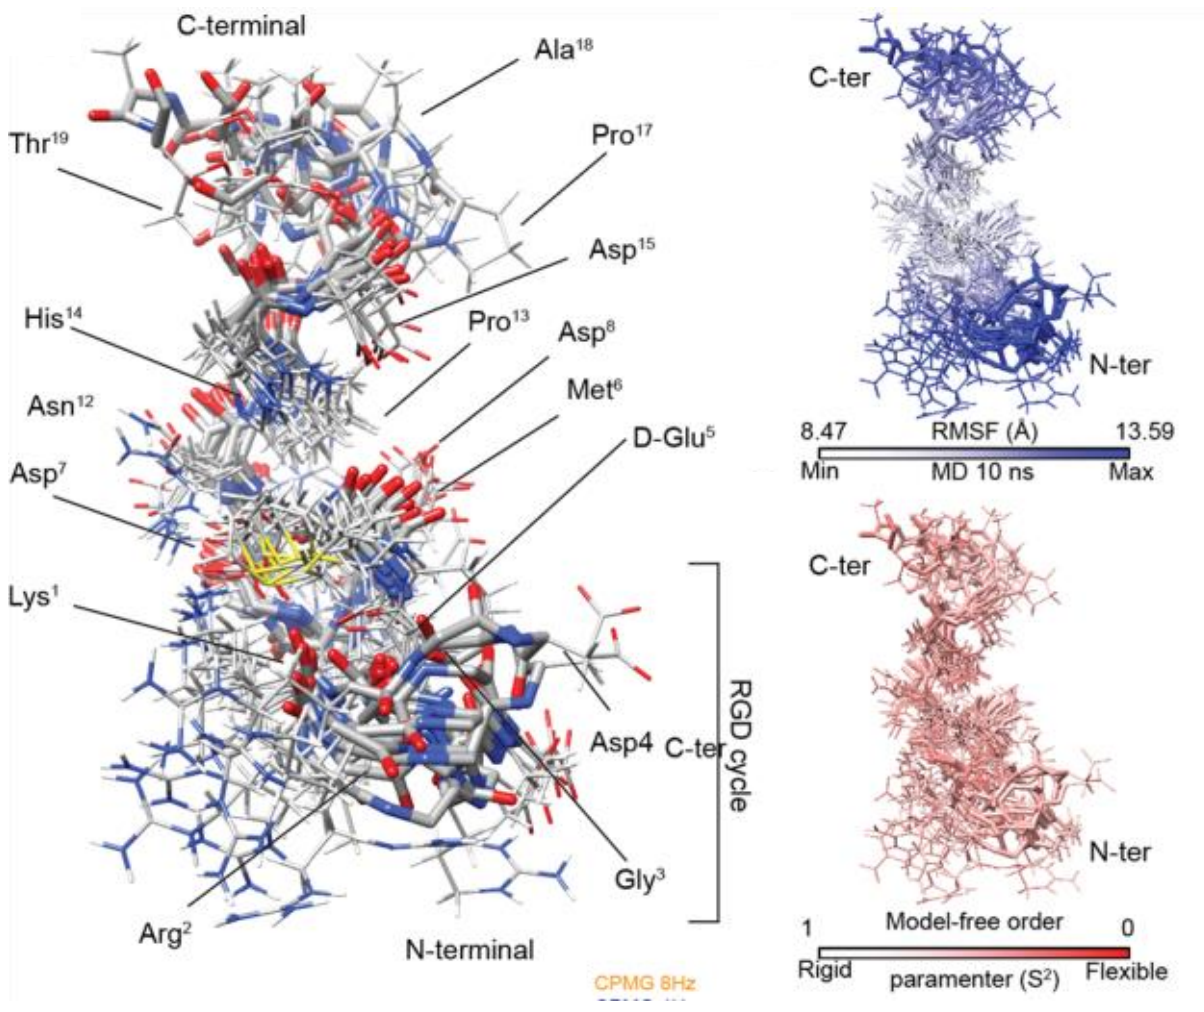
C-terminal
Ala18
Thr19
Pro17
Asp15
His14
Pro13
Asp8
Met6
Asn12
D-Glu5
Asp7
Lys1
Asp4
C-ter
RGD cycle
Gly3
Arg2
N-terminal
C-ter
N-ter
8.47
RMSF (Å)
13.59
Min
MD 10 ns
Max
C-ter
N-ter
1
Model-free order
0
Rigid
paramenter (S2)
Flexible

µs-ms dynamics of RGDechi15D by natural-abundance NMR spectroscopy

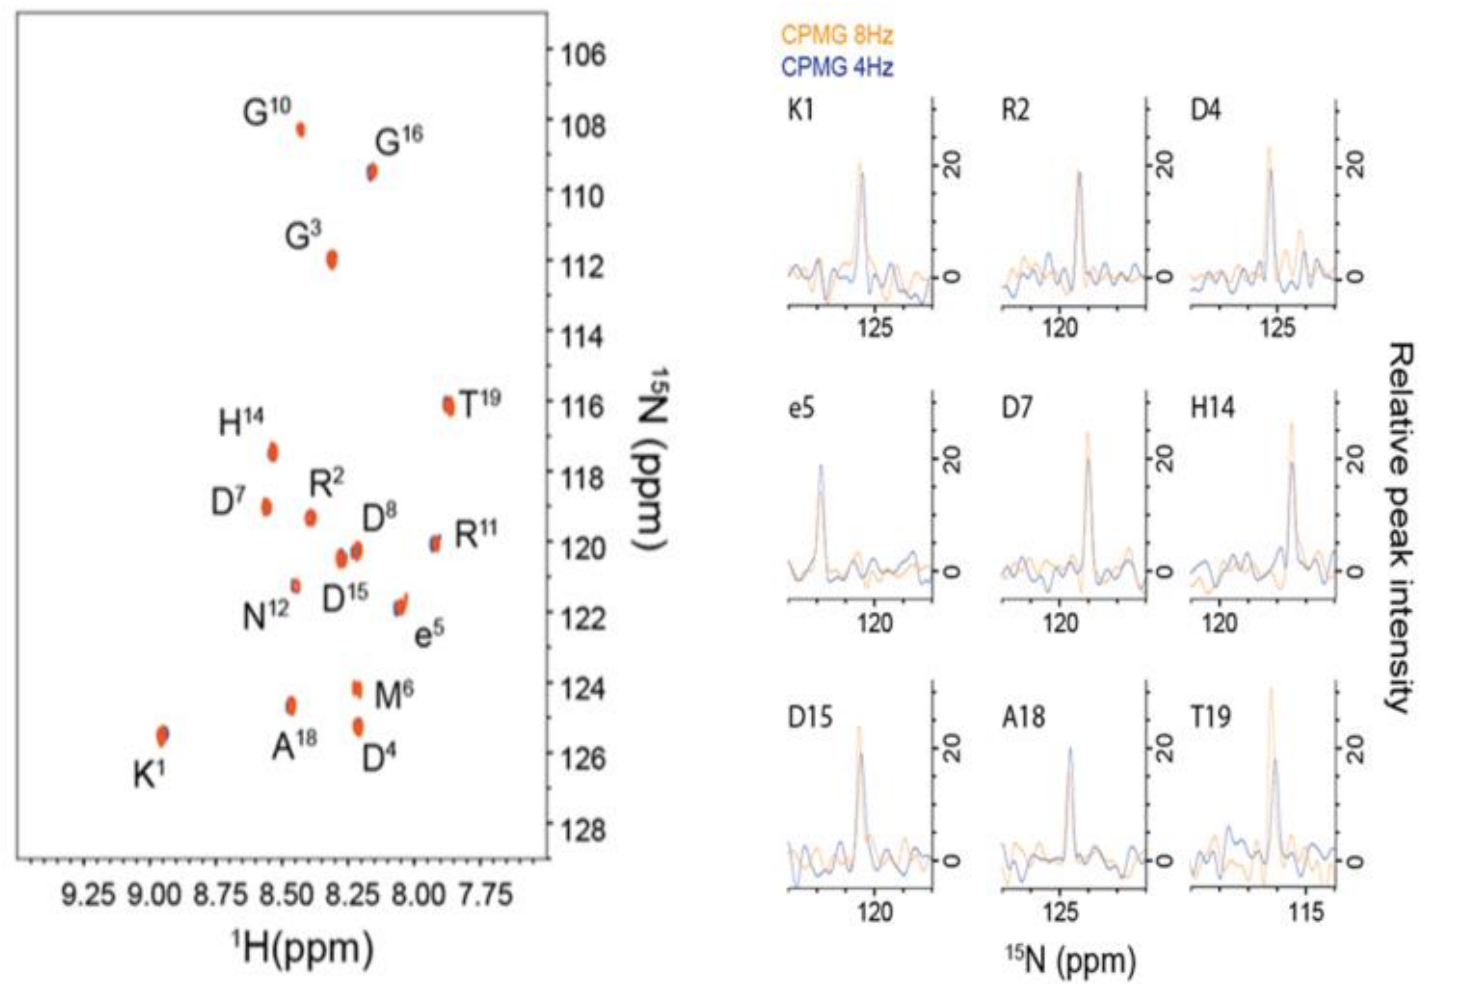
G10
G16
G3
H14
T19
D7
R2
D8
R11
N12
D15
e5
M6
K1
A18
D4
15N (ppm)
1H(ppm)
CPMG 8Hz
CPMG 4Hz
K1
R2
D4
e5
D7
H14
D15
A18
T19
Relative peak intensity
15N (ppm)

Mapping of the binding interface between RGDechi15D and αvβ5 integrin on living HeLa cancer cells surface

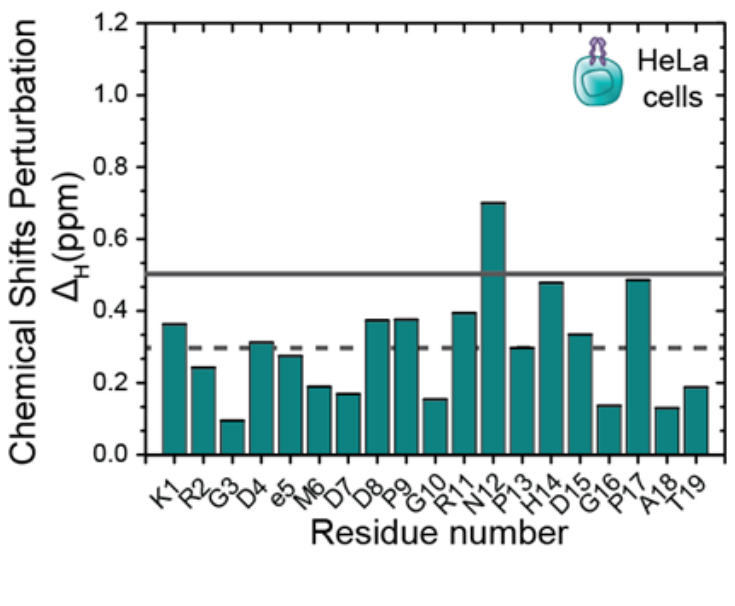
HeLa cells
Chemical Shifts Perturbation
ΔH(ppm)
Residue number

2D NUS trNOESY

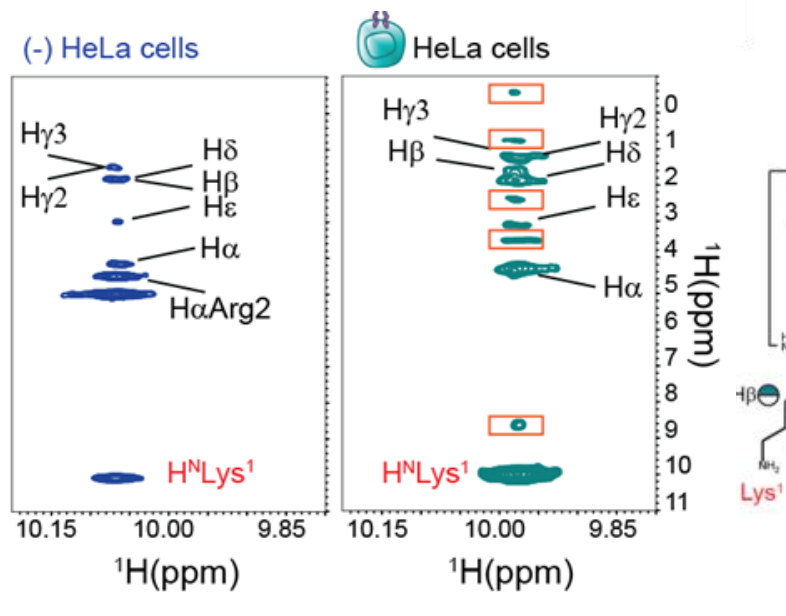
(-) HeLa cells
HeLa cells
Hγ3
Hγ2
Hδ
Hβ
Hε
Hα
HαArg2
HNLys1
1H(ppm)
1H(ppm)

1D T1ρ NMR experiments

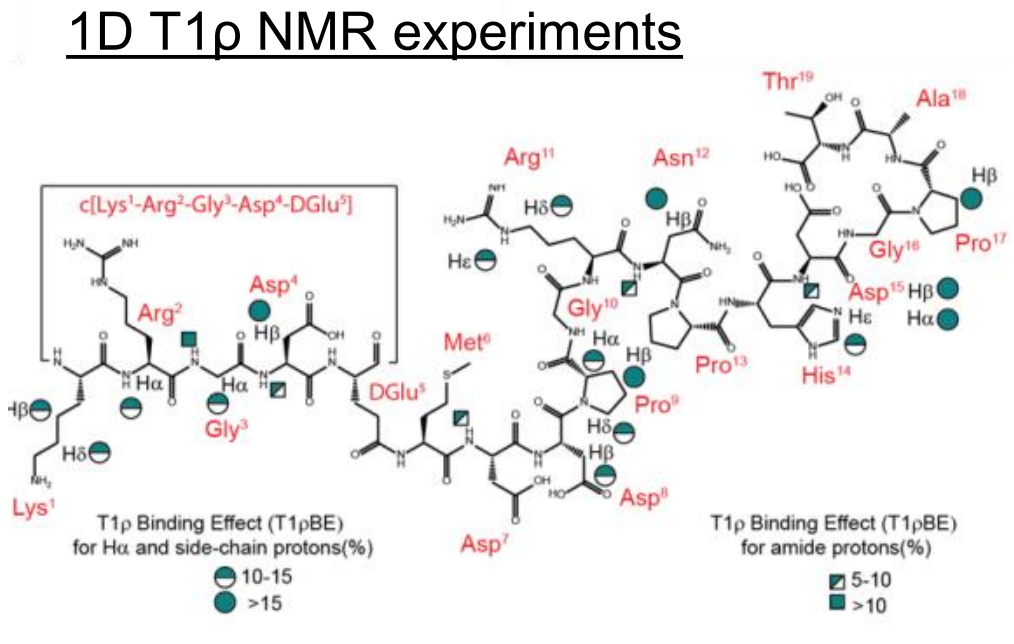
c[Lys1-Arg2-Gly3-Asp4-DGlu5]
T1ρ Binding Effect (T1ρBE) for Hα and side-chain protons(%)
10-15
>15
T1ρ Binding Effect (T1ρBE) for amide protons(%)
5-10
>10

The RGDechi15D/αvβ5 complex

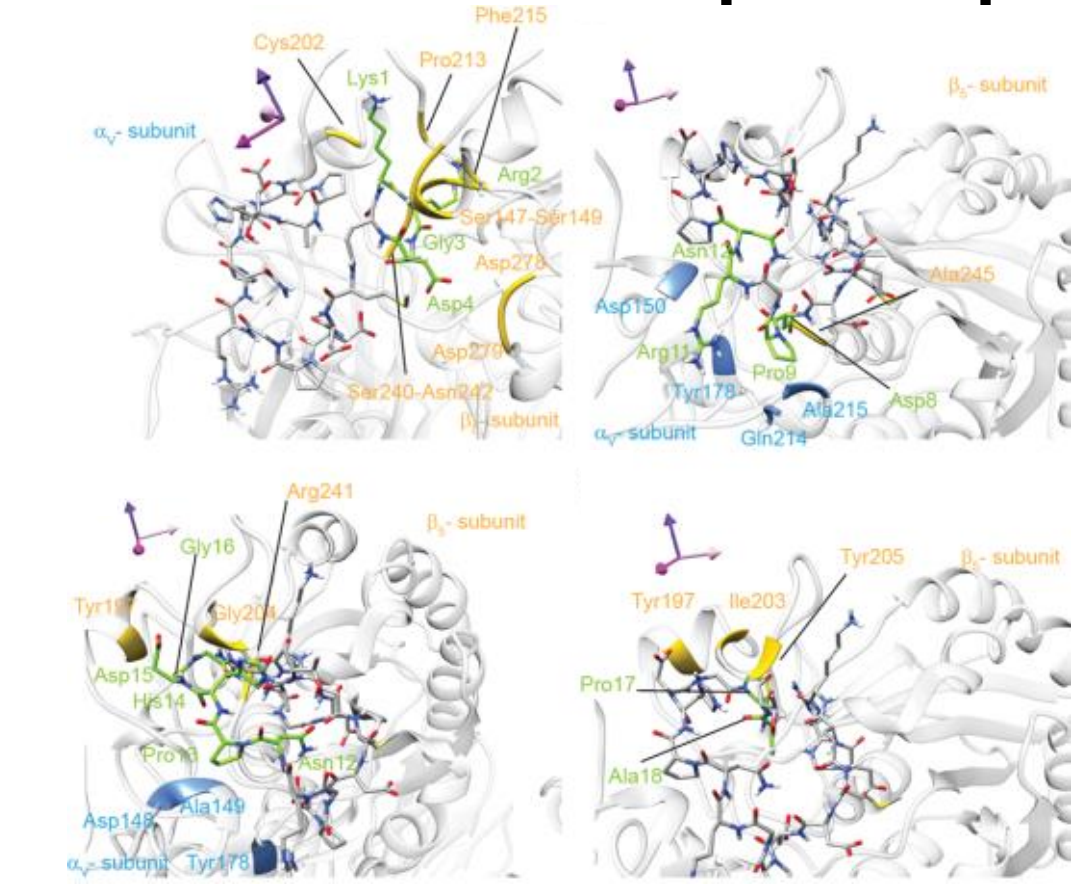
αV- subunit
β5- subunit

Conclusions

Herein, we developed and applied an alternative methodology for studying receptor-ligand interactions with living cells at atomic resolution. This application combines high-resolution structural and dynamics NMR data with Molecular Dynamics simulations and Molecular Docking studies. Our developed methodology was successfully applied in the investigation of the direct binding of the RGDechi15D peptide with the αvβ5 integrin with living human cancer cells. The structural model of RGDechi15D/αvβ5 complex indicates that the recognition mechanism is mainly mediated by the residues of the RGD motif and it is further stabilized by the residues located in the C-terminal region of the peptide. This approach represents an alternative NMR tool for studying, at atomic resolution, receptor-ligand recognition mechanism on living cells surface.

References

[1] E. Luchinat, L. Banci IUCrJ. 2017, 4, 108-18; [2] E. Luchinat, L. Banci J Biol Chem. 2016, 291, 3776-84; [3] B. Farina, I.de Paola, L. Russo, D. Capasso, A. Liguoro, A. Del Gatto, et al. Chemistry 2016, 22, 681-93; [4] D. Capasso, A. Del Gatto, D. Comegna, L. Russo, R. Fattorusso, M. Saviano, et al. Molecules 2020, 25.